CPL-410-005, A NOVEL UBIQUITIN-ACTIVATING ENZYME (UAE) INHIBITOR IN PRECLINICAL EVALUATION AS AN ANTICANCER TREATMENT FOR SOLID TUMOURS

D. Kozlowska[1], A. Górnicka[1], B. Stypik[2], M. Mroczkiewicz[2], A. Mikolajczyk[1,3], J. Hucz-Kalitowska[1], A. Szwalbe[2], K. Mulewski[2], D. Smuga[2], K. Dubiel[2], J. Pieczykolan[1], M. Wieczorek[4], A. Stanczak[1]

[1]Preclinical Research Dept., Celon Pharma S.A., Kiełpin/Łomianki, Poland; [2]Medicinal Chemistry Dept., Celon Pharma S.A., Kiełpin/Łomianki, Poland; [3]Postgraduate School of Molecular Medicine, Warsaw, Poland;
[4]Research and Development Center, Celon Pharma S.A., Kiełpin/Łomianki, Poland

corresponding author: aleksandra.stanczak@celonpharma.com; presentation number: PB-052

CELON PHARMA

Shape the future of healthcare.

INTRODUCTION

UPS (ubiquitin-proteasome system) is essential in cancer development and progression, since protein degradation is disturbed in comparison to non-malignant cells. This feature provides a new opportunity to target protein homeostasis and ubiquitin signalling for the treatment of cancer. Bortezomib is the first proteasome inhibitor approved by EMA and FDA for the blood cancer treatment. However, drugs beneficial for solid tumours are still missing. The strategy based on targeting the E1 enzyme (UAE) triggering UPS cascade may demonstrate clinical benefits in solid tumours. Currently, only MLN7243 (Takeda/Millenium) is in phase 1 clinical trial. CPL-410-005 compound was designed and developed as an E1 small-molecule inhibitor.

MATERIALS AND METHODS

Cell viability assay

Cells were seeded in 96-well plates and incubated for 72h with increasing doses of the compounds. Cell viability was assessed by ATPlite assay (Perkin Elmer) following the manufacturer's instructions. IC_{50} values were calculated with GraphPad Prism.

Western blot analysis

Cells were seeded in 6-well plates and treated with the compounds for 4h to assess ubiquitylation, SUMOylation and neddylation levels, or for 24h to examine UPR and apoptosis. Then, cells were lysed and examined by Western blotting, according to the protocols provided by the antibody suppliers. The antibodies against NEDD8, ubiquitin and SUMO2/3/4 were purchased from Santa Cruz Biotechnology. Antibodies against IRE1, BiP, PERK, PARP and caspase 3 were obtained from Cell Signaling Technology.

Microsomal stability

Microsomes were incubated with CPL-410-005 and MLN7243 with or without appropriate cofactors. The reaction was stopped in consecutive time points and subsequently samples were analysed by LC/MS.

Solubility and Lipophilicity

Solubility and distribution coefficients logD have been determined using the shake-flask technique in buffers or biphasic octanol-buffer mixtures, respectively. The pH values of the buffers used in the assay correspond to the acidity along the GI tract. The compounds were quantitatively determined by HPLC or LC-MS techniques.

Genotoxicity

To analyse the mutagenicity of CPL-410-005, Ames MPFTM test (Xenometrix AG) with *Salmonella typhimurium* strain TA98 in the presence/absence of rat liver S9 fraction was performed.

PK/PD animal study

HCT-116 ($3x10^6$ cells per animal) cells were mixed with PBS/Matrigel and injected subcutaneously into SCID mice (Charles River). Mice were randomized into groups (n=24) when tumours reached the determined size of more than 100 mm^3. A single-dose of 5mg/kg body weight CPL-410-005 or MLN7243 was administered intravenously. At indicated timepoints, blood and tumours were collected (4 animals per timepoint). The plasma and tumour concentration-time profiles of test compounds were analysed using UHPLC-MS.

REFERENCES

[1] Nalepa *et al*. Drug discovery in the ubiquitin-proteasome system. *Nature* (2006) 5, 596-613.
[2] Yang *et al*. Inhibitors of Ubiquitin-Activating Enzyme (E1), a New Class of Potential Cancer Therapeutics. *Cancer Res* (2007) 67, 9472-9481.
[3] Xu *et al*. The ubiquitin-activating enzyme E1 as a therapeutic target for the treatment of leukemia and multiple myeloma. *Blood* (2010) 115, 2251-2259.
[4] Ungermannova *et al*. Identification and Mechanistic Studies of a Novel Ubiquitin E1 Inhibitor. *J Biomol Screen* (2012) 17, 421-434.

RESULTS

CPL-410-005 inhibits cell viability of tumour cells with greater potency than MLN7243

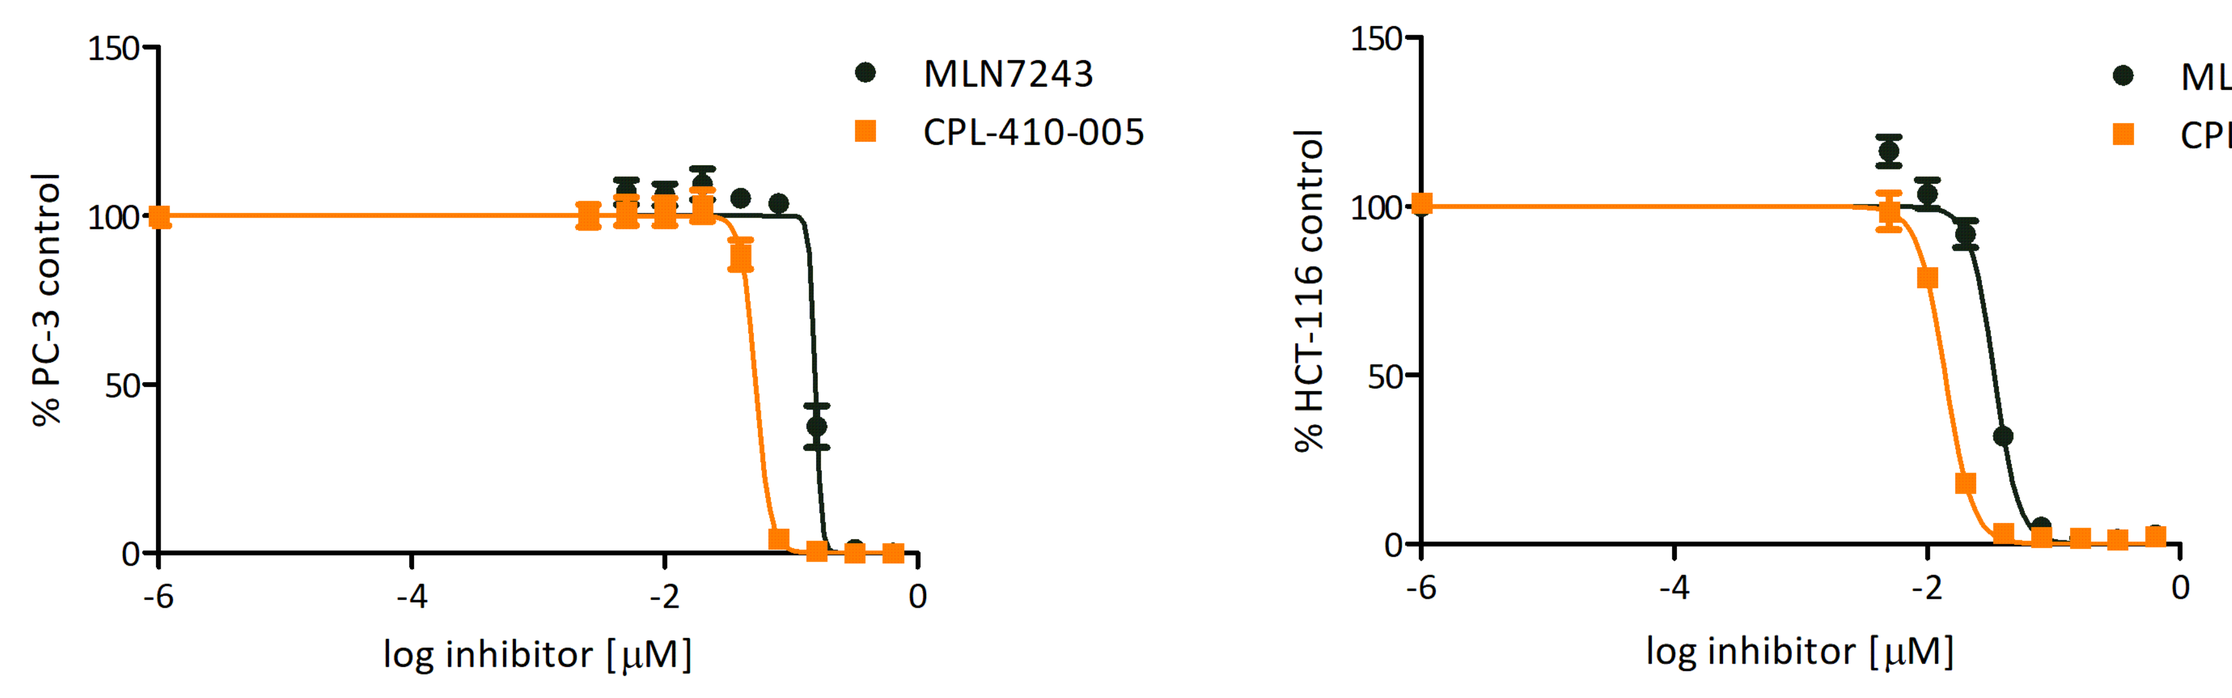
% PC-3 control
log inhibitor [μM]
% HCT-116 control
log inhibitor [μM]
MLN7243
CPL-410-005

CPL-410-005 is a more potent inhibitor of cell viability of colon cancer cells (HCT-116) and prostate cancer cells (PC-3) at low nanomolar IC_{50} (13.8 nM and 52.7 nM, respectively), than reference compound MLN7243 (40.0 nM and 155.6 nM, respectively).

CPL-410-005 inhibits cellular polyubiquitylation, but not neddylation/ SUMOylation

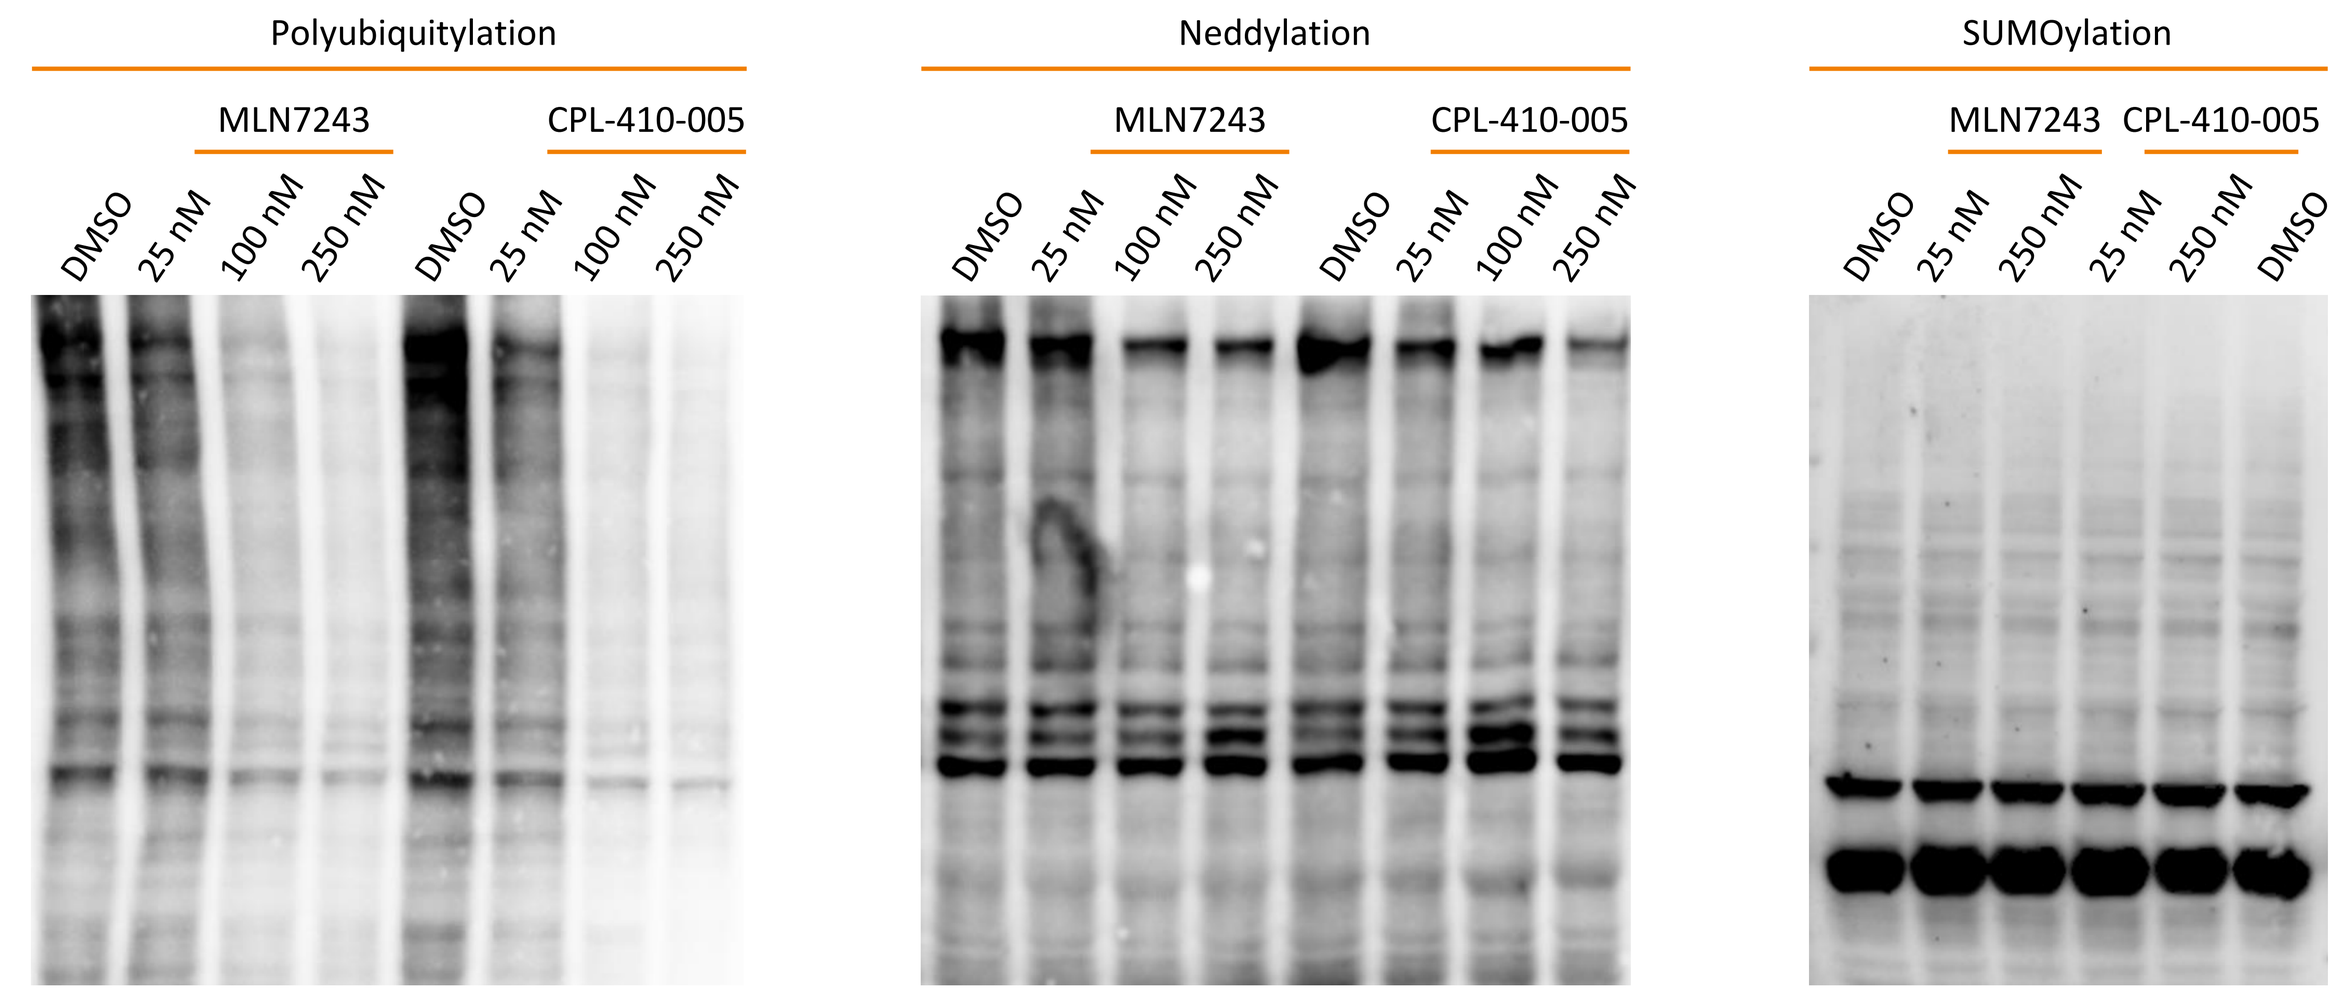
Polyubiquitylation
Neddylation
SUMOylation
MLN7243
CPL-410-005
DMSO
25 nM
100 nM
250 nM

In colorectal cancer cells (HCT-116), polyubiquitylation, but not neddylation nor SUMOylation, was inhibited after 24h of treatment of CPL-410-005.

Unfolded protein response and programmed cell death were induced after CPL-410-005 treatment

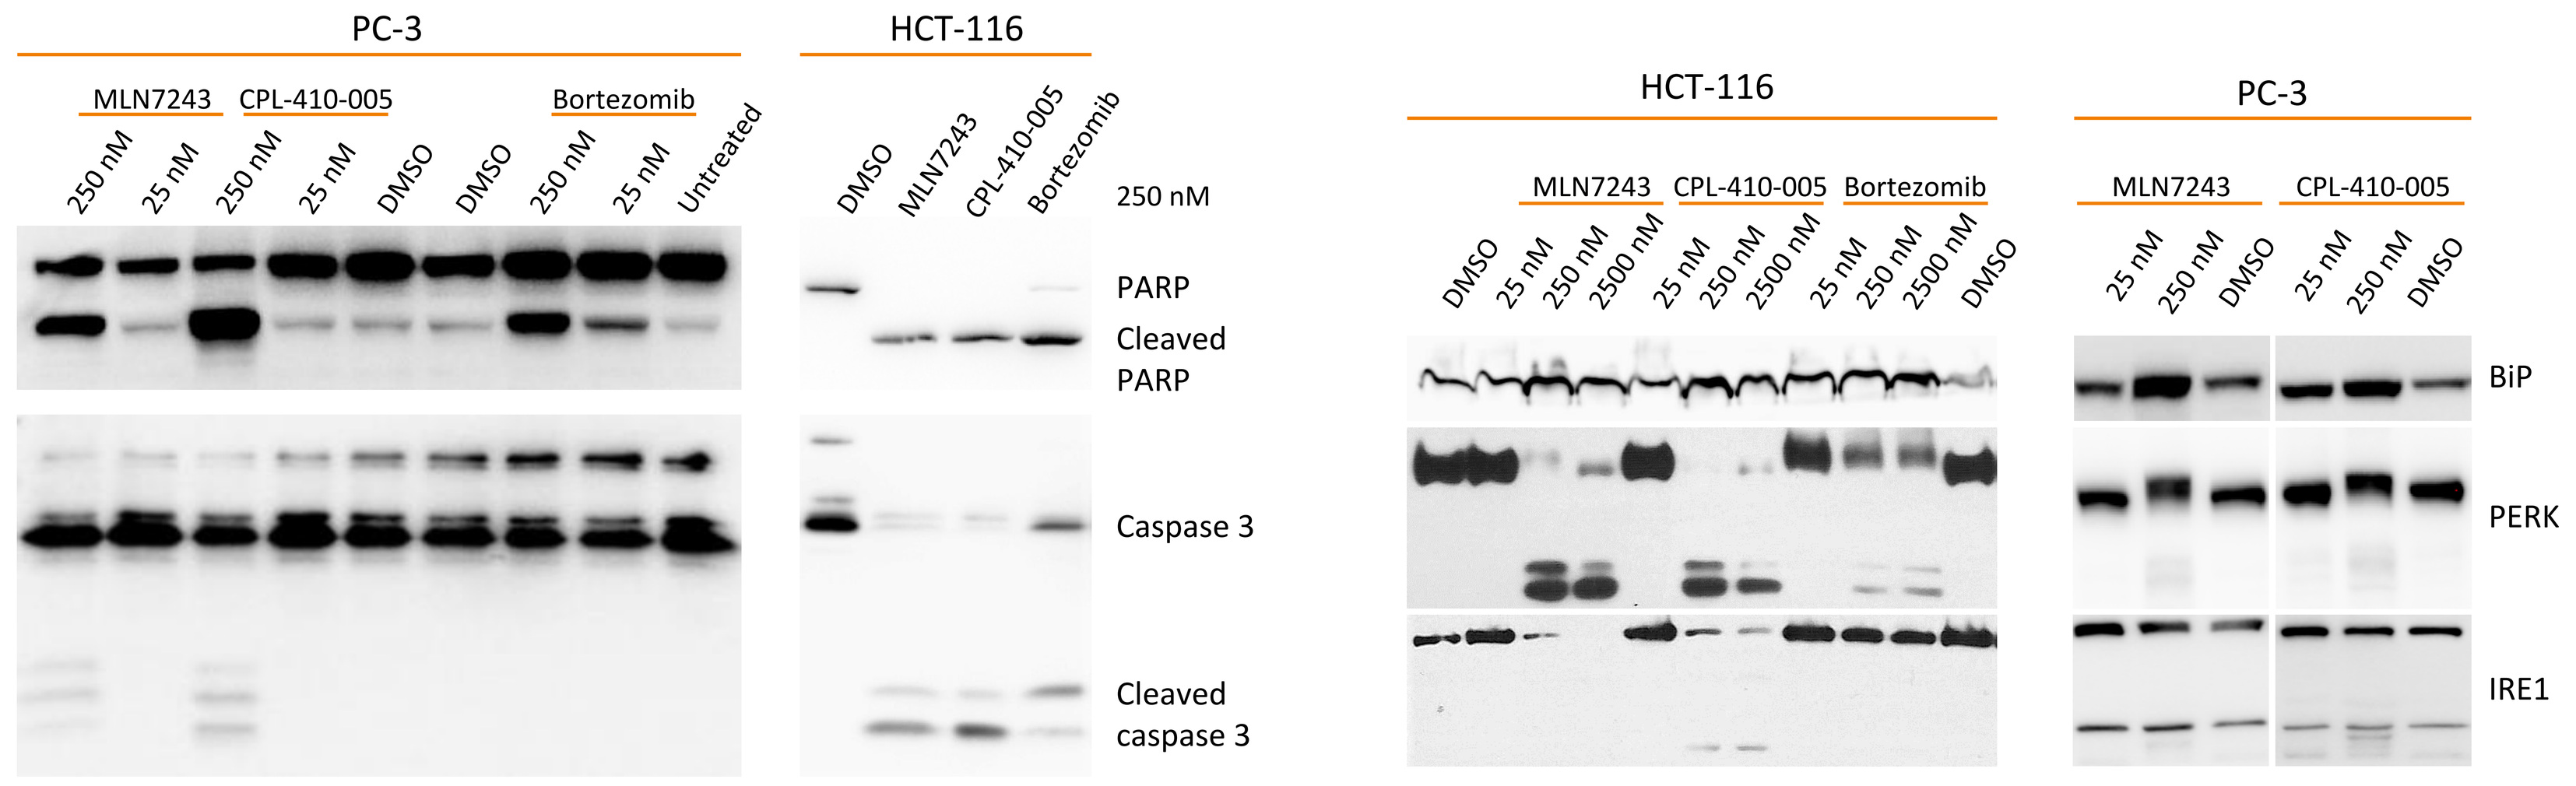
PC-3
HCT-116
MLN7243
CPL-410-005
Bortezomib
Untreated
DMSO
250 nM
PARP
Cleaved PARP
Caspase 3
Cleaved caspase 3
BiP
PERK
IRE1

In colorectal cancer cells (HCT-116) and prostate cancer cells (PC-3), PARP and caspase-3 were cleaved after 24h of treatment with CPL-410-005. Moreover, CPL-410-005 treatment of HCT-116 and PC-3 cells led to unfolded protein response by increased phosphorylation of PERK and IRE-1 (band shift) and BiP expression.

European Union
European Regional Development Fund
The National Centre for Research and Development
European Funds
Smart Growth
This study was supported by The National Centre for Research and Development (2/1.2/2017).

RESULTS

CPL-410-005 exhibits drug-like properties – low lipophilicity, high metabolic stability, no mutagenicity

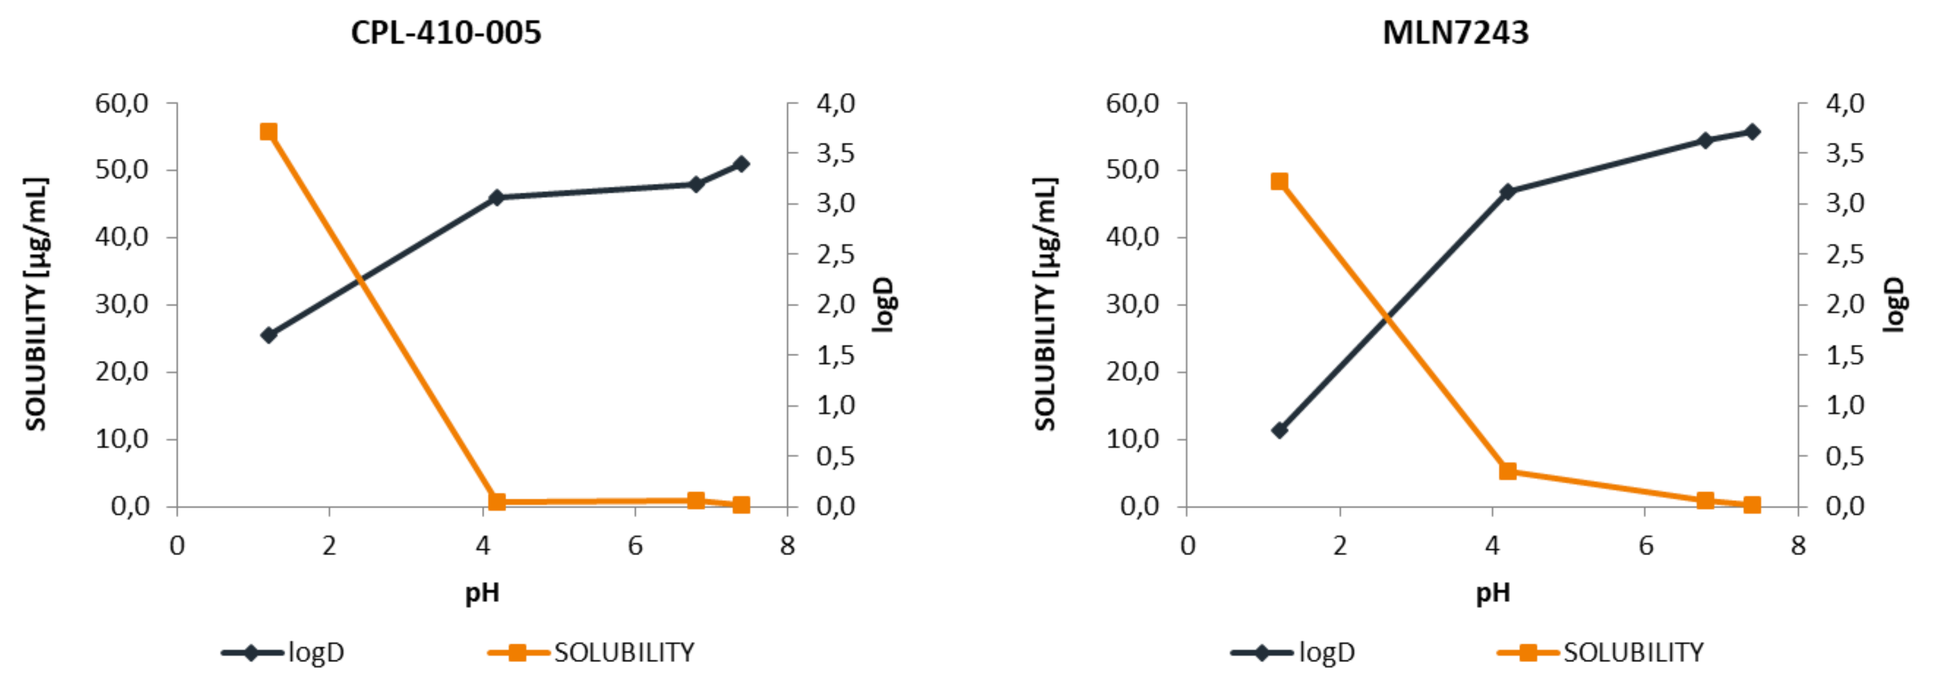
CPL-410-005
MLN7243
SOLUBILITY [μg/mL]
logD
pH
logD
SOLUBILITY

The solubility (0.3–55 μg/mL) and lipophilicity of CPL-410-005 ranges 1.7–3.4 and is comparable to MLN7243.

	Microsomal stability			
	Microsomes - murine		Microsomes - human	
Compound	T1/2 [min]	Cl [μ/min*mg]	T1/2 [min]	Cl [μ/min*mg]
MLN7243	74	18,6	362	3,8
CPL-410-005	294	4,7	493	2,8
Donepezil	51	27	699	2
7-HFC	3	461	3	405

CPL-410-005 exhibits higher metabolic stability on mice and human in comparison to MLN7243. The obtained data was referred to the compounds of known stability parameters (Donepezil, 7-HFC: 7-Hydroxy-4-(trifluoromethyl)coumarin).

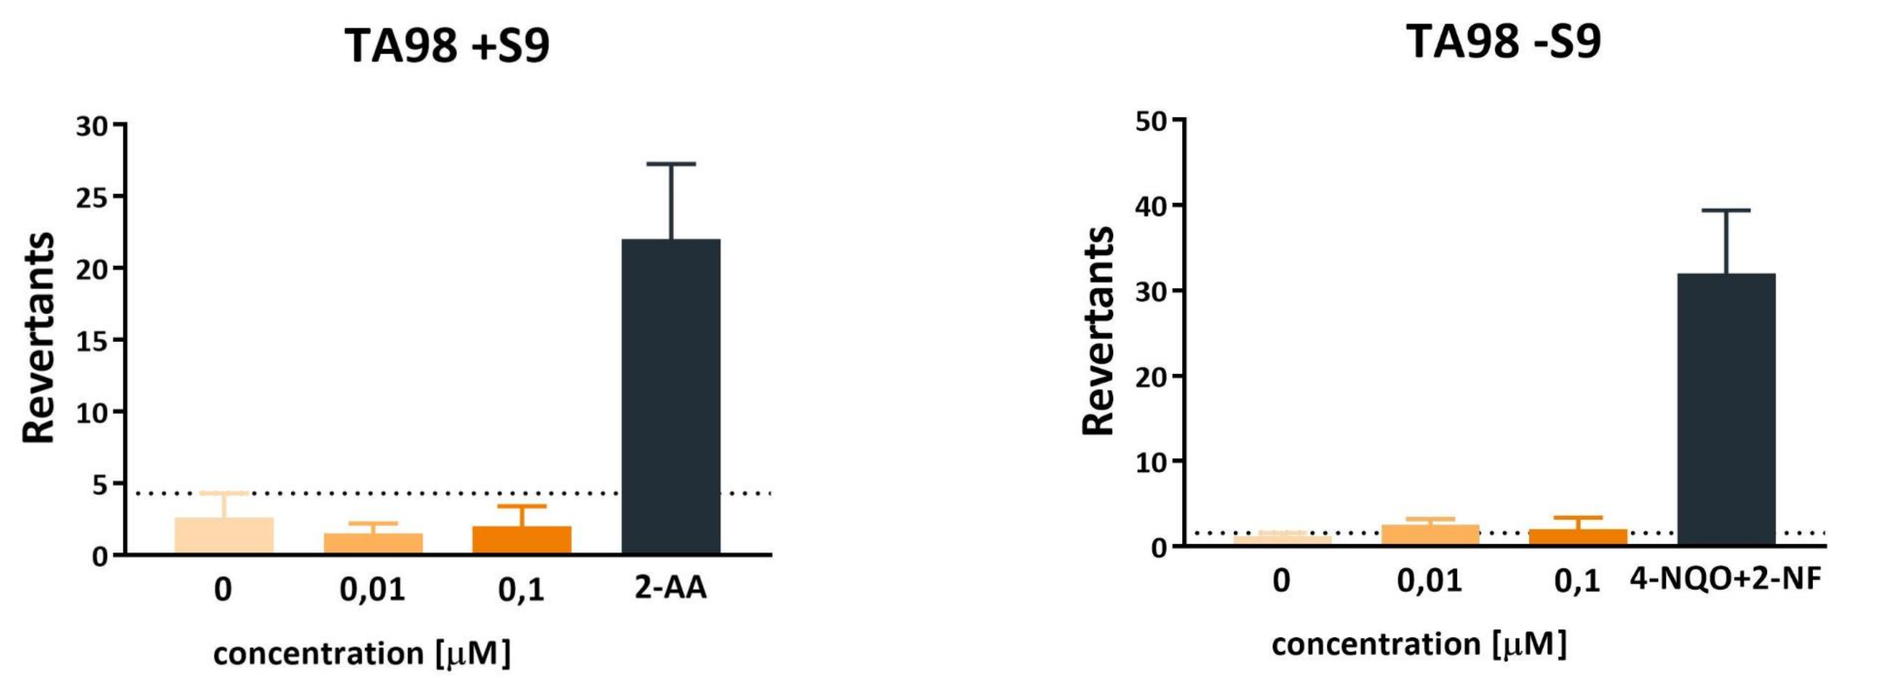
TA98 +S9
TA98 -S9
Revertants
concentration [μM]
0
0,01
0,1
2-AA
4-NQO+2-NF

Number of revertants of *Salmonella typhimurium* TA98 treated with CPL-410-005 was below the base level, either in the presence or absence of rat liver S9 fraction. The obtained data was referred to the compounds of known genotoxic potential (2-NF: 2-Nitrofluorene; 4-NQO: 4-Nitroquinoline-N-oxide; 2-AA: 2-Amino-anthracene).

CPL-410-005 is rapidly eliminated from the plasma but retained in tumour

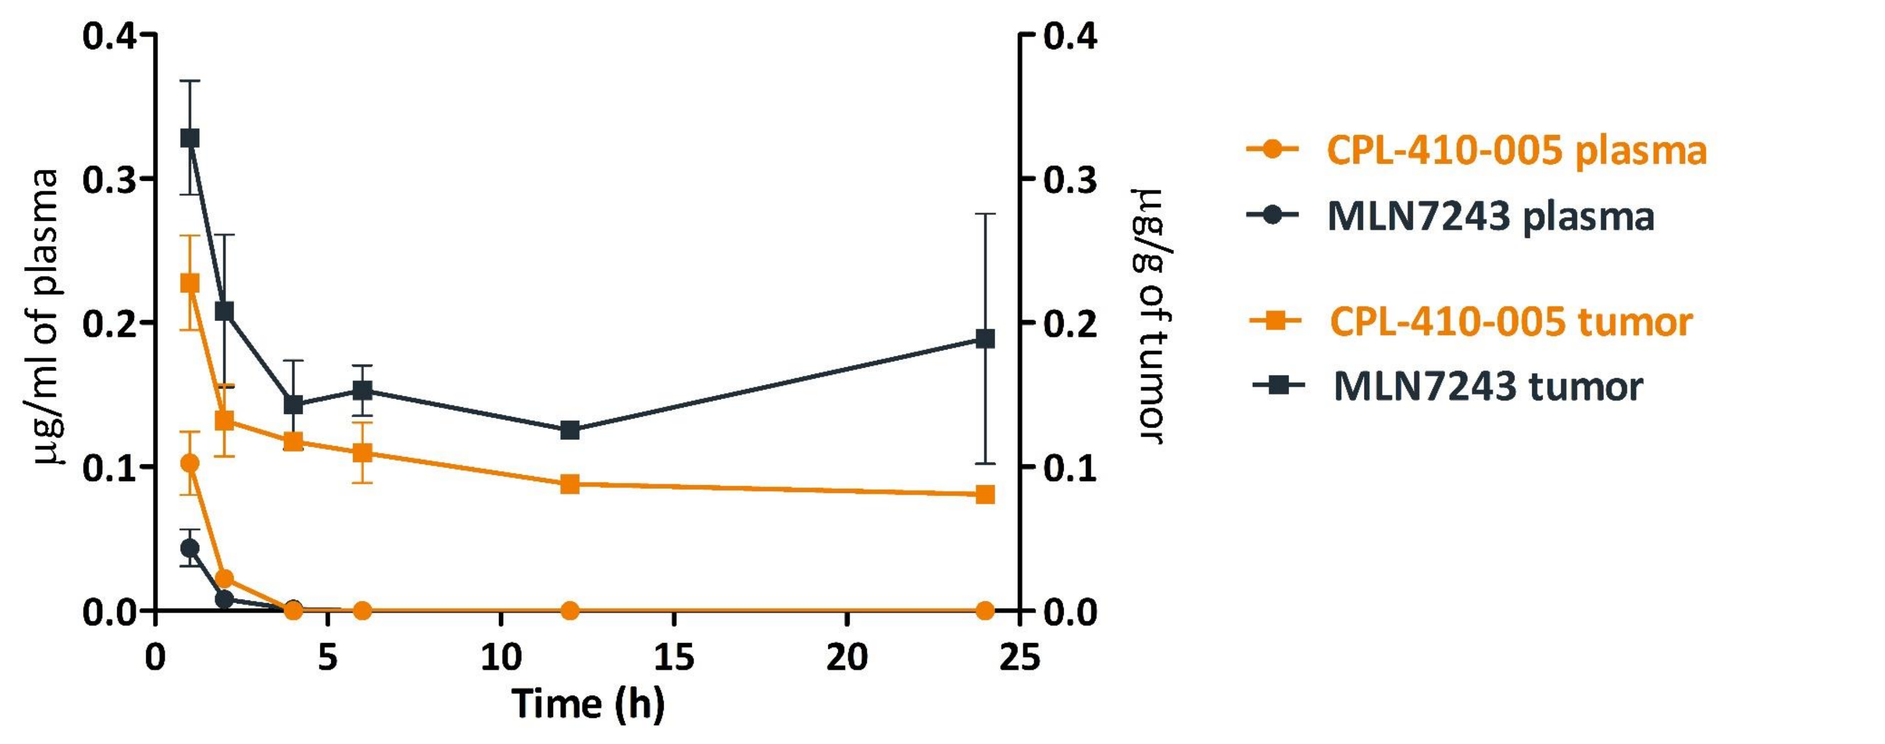
μg/ml of plasma
μg/g of tumor
Time (h)
CPL-410-005 plasma
MLN7243 plasma
CPL-410-005 tumor
MLN7243 tumor

In SCID mice bearing human tumour HCT-116 xenografts, single IV dosing of 5mg/kg CPL-410-005 has shown rapid elimination from the plasma and long retention in tumour (up to 24h).

CONCLUSIONS

We have designed a potent E1 inhibitor with promising *in vitro* and *in vivo* results. Preclinical findings may lead to the development of a novel anticancer therapy for solid tumours.