Development of SR matrix tablet containing a new GPR40 agonist for first-in-human clinical trial

Ewelina Juszczyk[1], Kamil Kisło[1], Paweł Żero[1], Ewa Tratkiewicz[1],
Jadwiga Paszkowska[2], Dorota Danielak[4], Jaroslaw Sczodrok[3], Grzegorz Garbacz[2,3]

[1] Celon Pharma S.A., Główna 48, 05-152 Kazuń Nowy, Poland, ewelina.juszczyk@celonpharma.com
[2] Physiolution Polska Sp. z o.o., Piłsudskiego 74, 50-020 Wrocław, Poland
[3] Physiolution GmbH, Walther-Rathenau-Str. 49 a, 17489 Greifswald, Germany
[4] Department of Physical Pharmacy and Pharmacokinetics, Poznan University of Medical Sciences, Święcickiego 6, 60-781, Poznań

CELON PHARMA

PHYSIOLUTION
Predictive Dissolution Testing

Purpose

We present development of MR tablets with a new antidiabetic agent GPR40 agonist for first-in-human administration in phase I clinical trial. The aim was to develop MR tablets containing 5, 30 or 120 mg of the drug that are resistant to the GI passage conditions in either fasted or fed states and assure same drug delivery kinetics regardless the dose labelled. The tablets should bear no risk of dose dumping and provide stable drug release over 14-18 hours. The aim of the formulation development was to minimize the risk and extend the duration of the therapeutic drug plasma levels.

Introduction

The development of oral dosage forms for the first-in-human (FIH) study of innovative medicines requires simple and reliable formulations that allow to estimate the bioavailability and in rare cases to achieve the desired drug delivery rate. In such cases the determination of simple oral solution or IR capsule is not necessarily recommended. This holds true especially if the API underlays a rapid elimination and long lasting exposition is required for the clinically desired pharmacodynamical effect and compliance. In such cases the development of MR products of reliable drug delivery performance should be taken into consideration.

When MR dosage form is rational for the delivery of a new drug in first-in-human administration, it is even more difficult to predict its performance in vivo. It is due to the fact that on the drug discovery stage there is still no human data that could give any hint regarding the PK properties of the studied medicine. Moreover, the use of standard dissolution test is not sufficient to evaluate in vivo behavior of the developed formulations [2]. In this case the bio-predictive dissolution devices are of great support and provide realistic estimation of the bio-pharmaceutical performance and physiology-driven formulation development [1].

Results

Excipients selection step
First stage of SR tablets development was to evaluate the performance of prototype compositions. During this step key factors influencing drug release were identified. First of all, the excipients compatible with the drug substance were selected and divided into two groups: matrix forming agents and filling agents. The former assured sustained release of a new API; the latter served as fillers responsible for appropriate tablet compression. Exemplary dissolution profiles of tested formulations are presented in Fig. 2.

Drug release in stress test device
Selected prototype formulations of best in vitro behavior were then analyzed in stress test device that allow simulation of fasted and fed conditions in biorelevant media and simulation of mechanical stress of physiological intensity, realistic transit times and pH. The differences in dosage form performance between the tested formulations were observed. Unwanted effects, such as too slow drug release or dose dumping upon mechanical agitation could be identified (Fig. 3). Consequently, optimization of the formulation resistant to GI tract conditions (fasted or fed state) and of desired release time was enabled. The results for 120 mg tablet strength are shown in Fig. 4

Development of common dissolution rate for three SR tablet strengths
The last stage of SR tablet development was to obtain an in vitro dissolution rate that would be independent of tablet strength administered to the patient. For this reason a common dissolution profile for all tablet strengths were aimed both in fasted and fed state. The performance of dosage forms was evaluated again in stress test device. Based on the results, relevant compositions that ensures similar dissolution behavior for three tablet strengths were selected for clinical study. Despite big differences in tablet strengths and thus, with differing composition of other excipients, a 100% dissolution was achieved in ca. 12-18 hours both in fasted and fed state, which was a clinically desired release time. The results from both states are presented in Fig. 5-6.

Figures

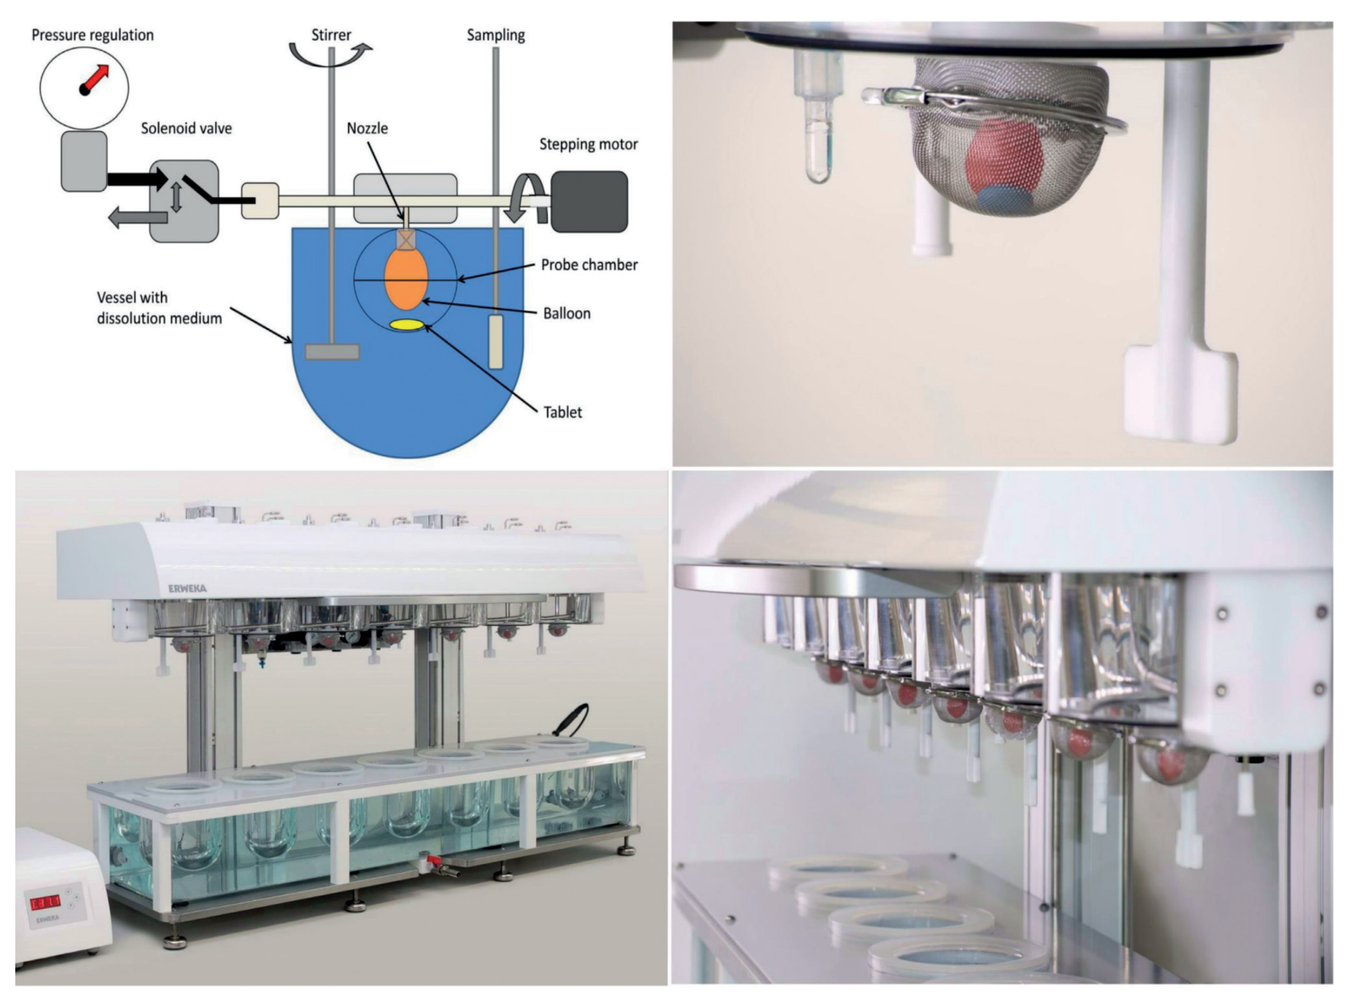
Pressure regulation
Stirrer
Sampling
Solenoid valve
Nozzle
Stepping motor
Probe chamber
Balloon
Vessel with dissolution medium
Tablet

Fig. 1: Scheme and picture of the dissolution stress test device

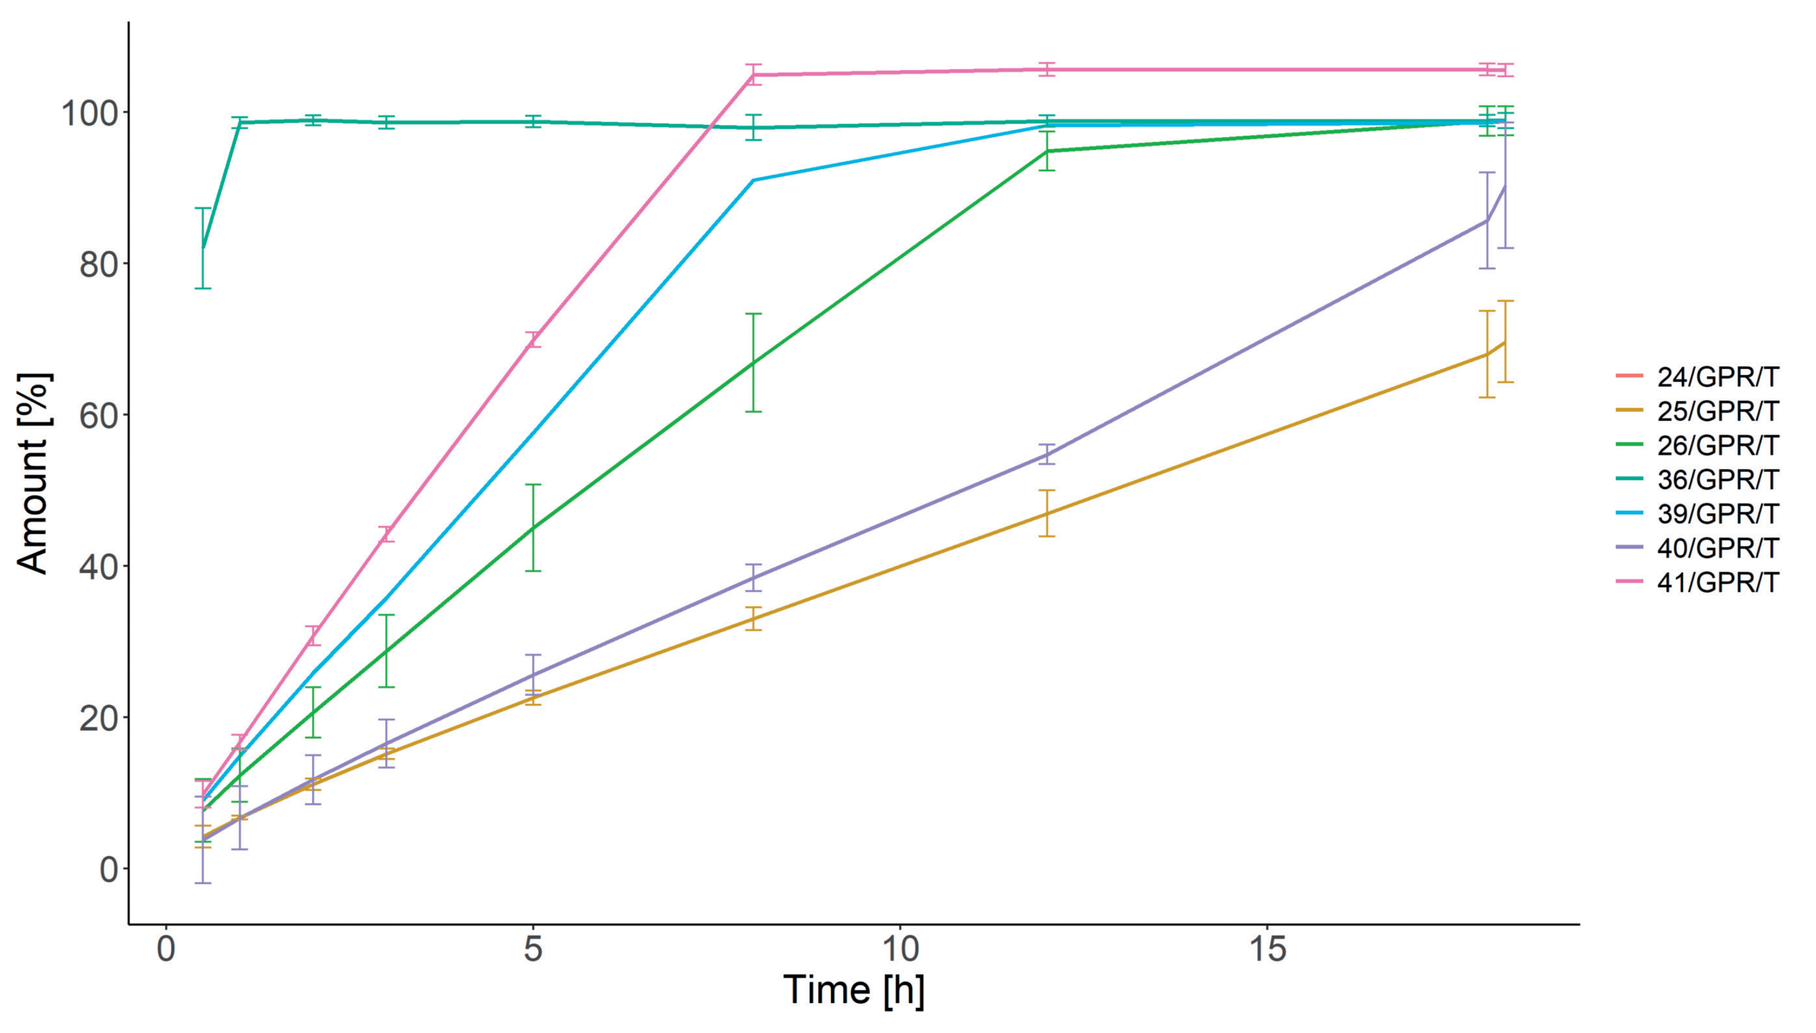
Amount [%]
Time [h]
24/GPR/T
25/GPR/T
26/GPR/T
36/GPR/T
39/GPR/T
40/GPR/T
41/GPR/T

Fig. 2: Mean dissolution profiles of formulations containing different amounts of matrix former, USP apparatus 2, 75 rpm, pH 6.8 phosphate buffer

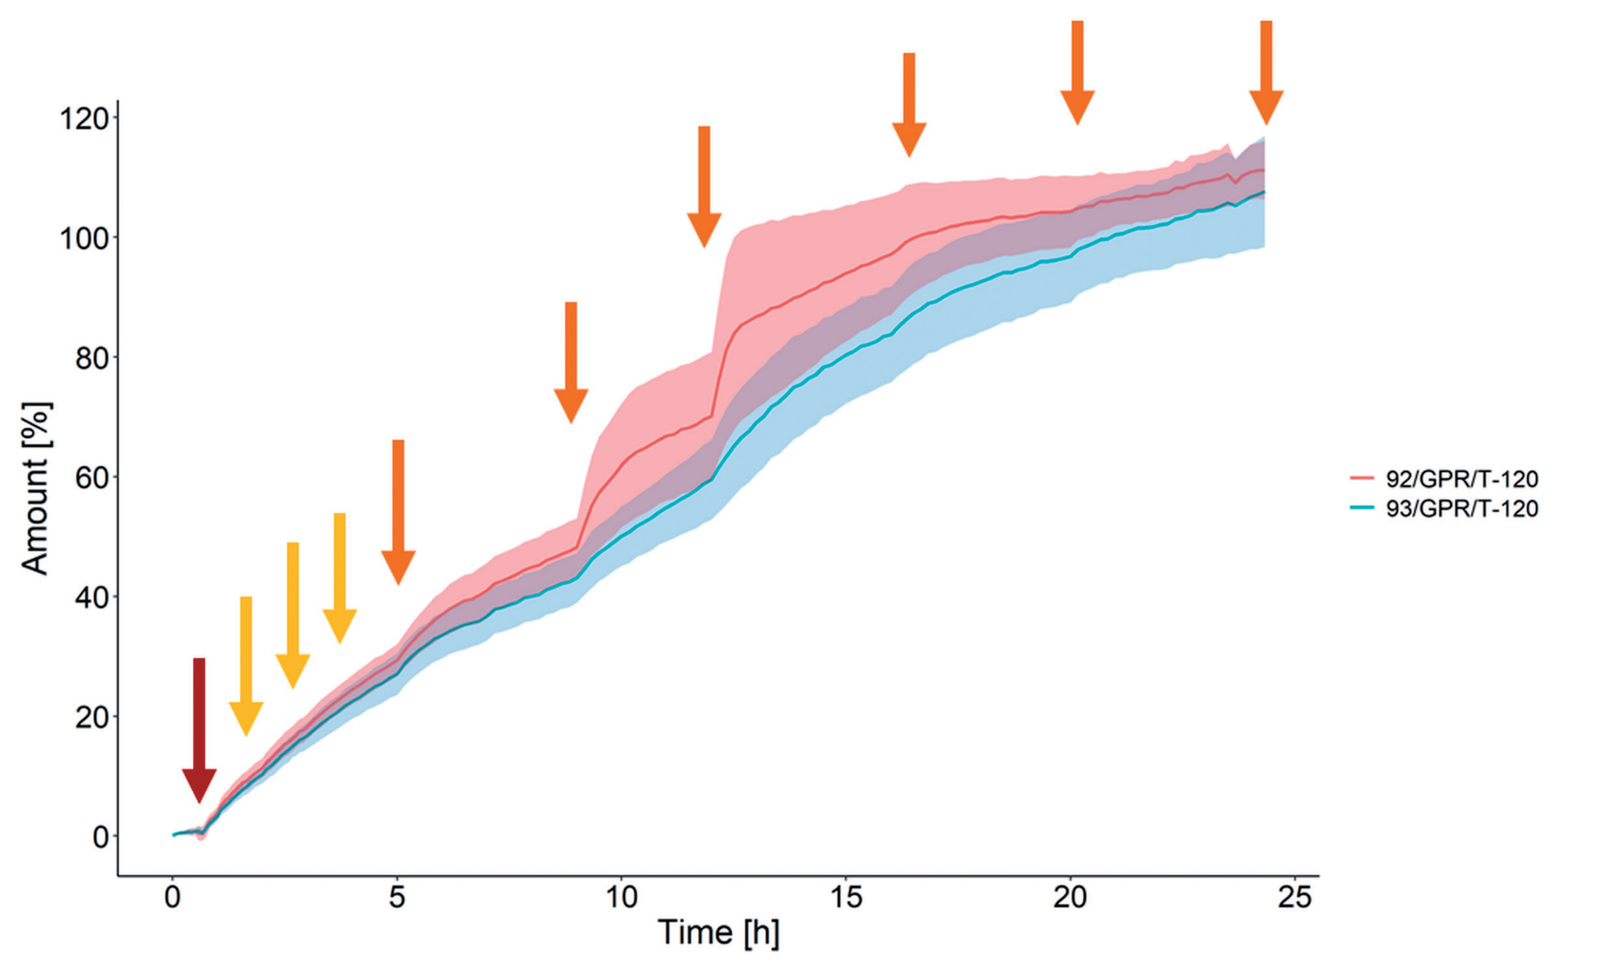
Amount [%]
Time [h]
92/GPR/T-120
93/GPR/T-120

Fig. 3: Identification of dose dumper in dissolution stress test

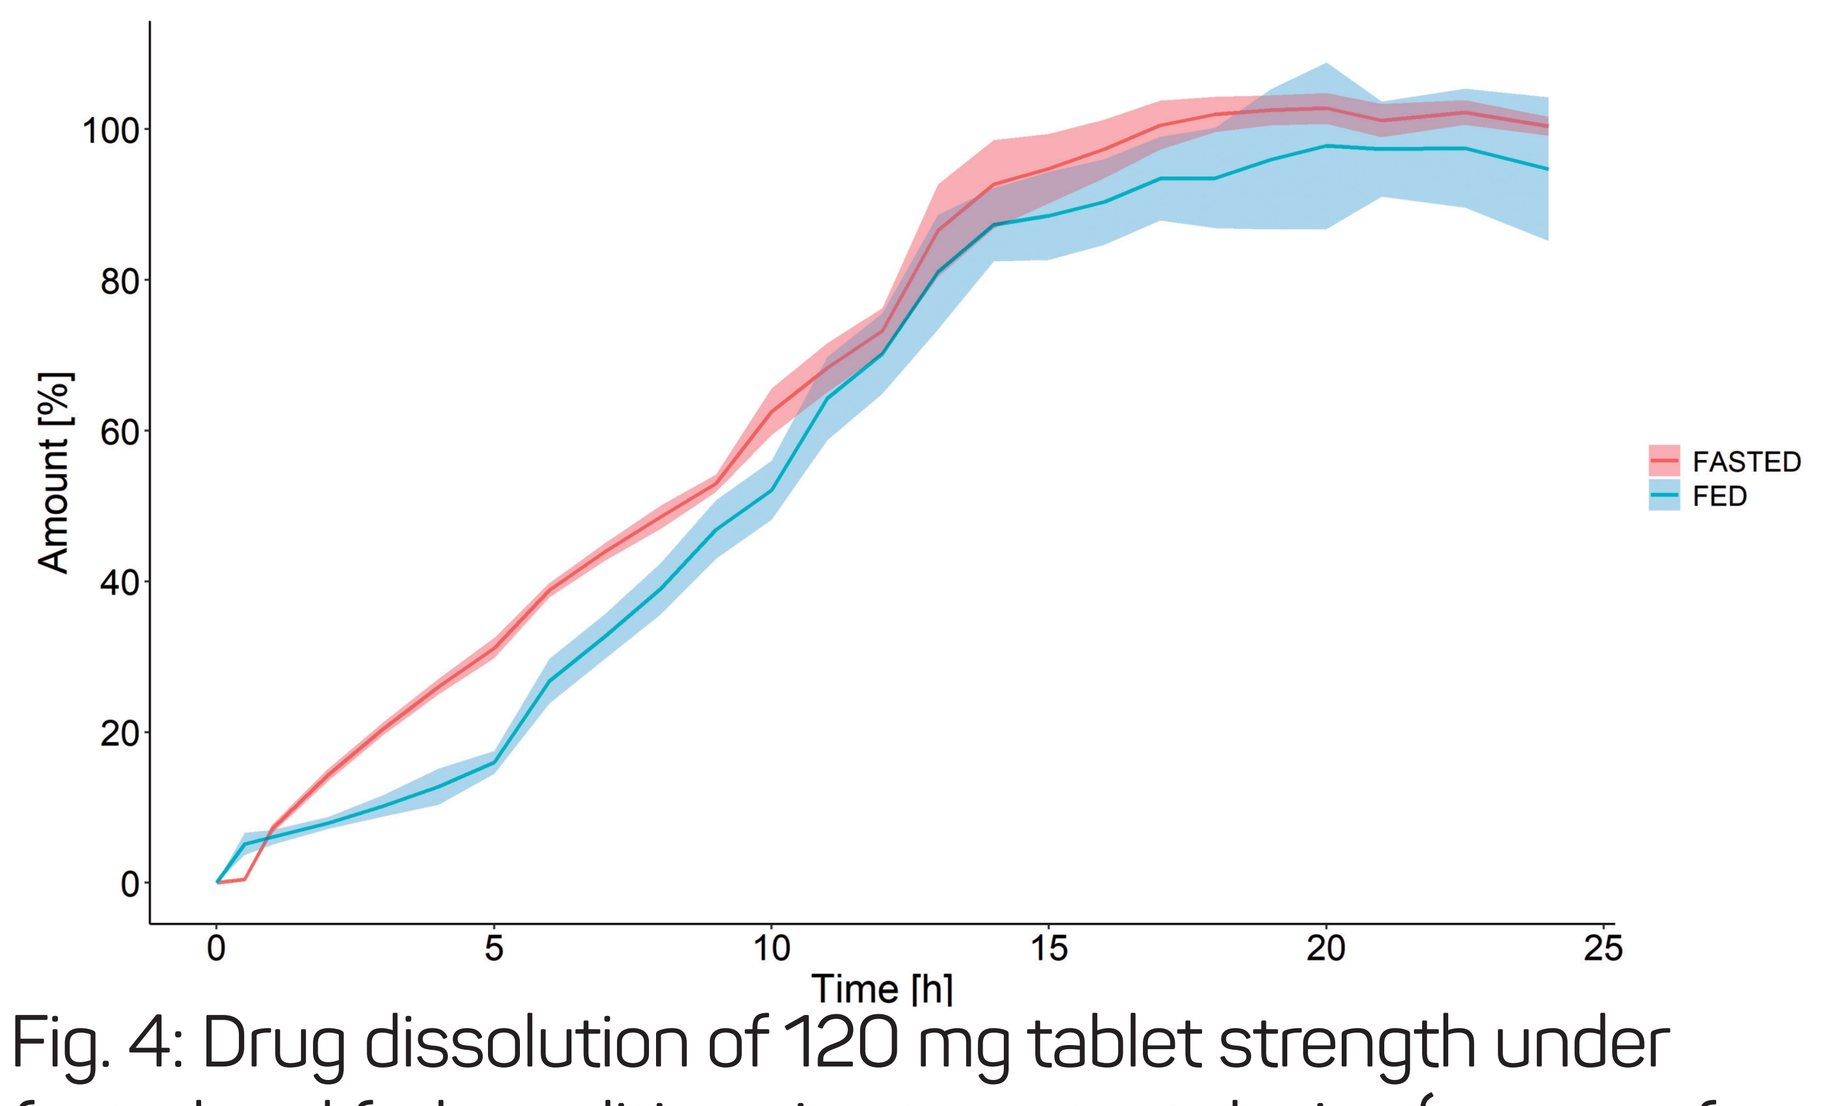
Amount [%]
Time [h]
FASTED
FED

Fig. 4: Drug dissolution of 120 mg tablet strength under fasted and fed conditions in stress test device (means of n=6±SD)

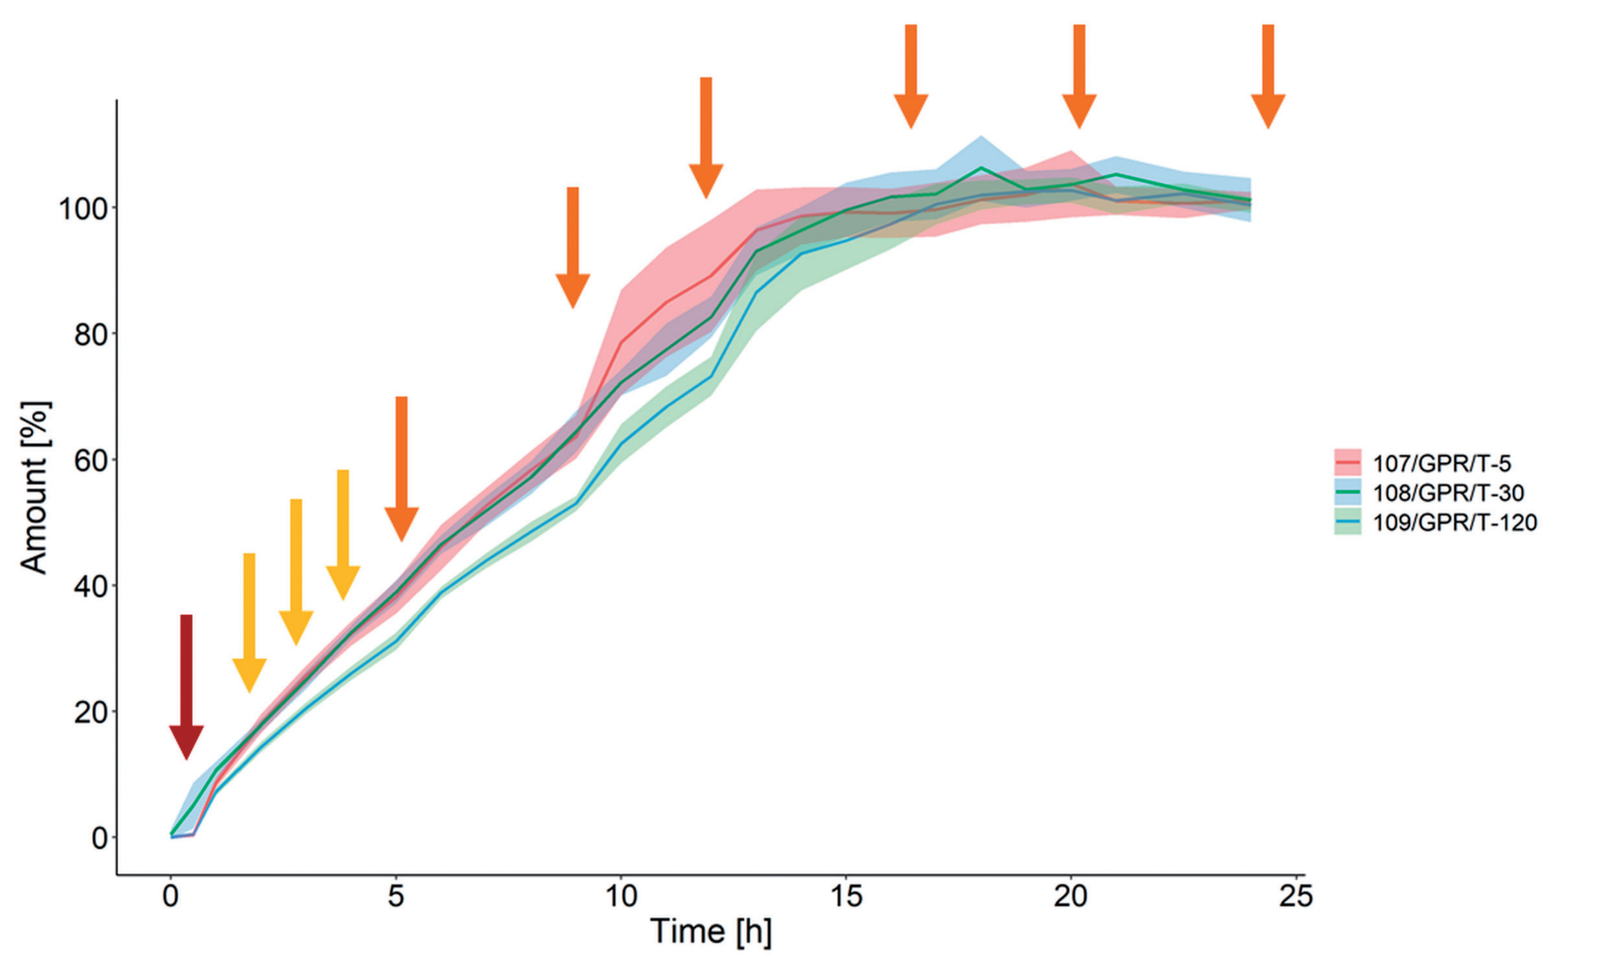
Amount [%]
Time [h]
107/GPR/T-5
108/GPR/T-30
109/GPR/T-120

Fig. 5: Dissolution profiles of three tablet strengths analyzed in stress test device under simulated fasted conditions (means of n=6±SD).

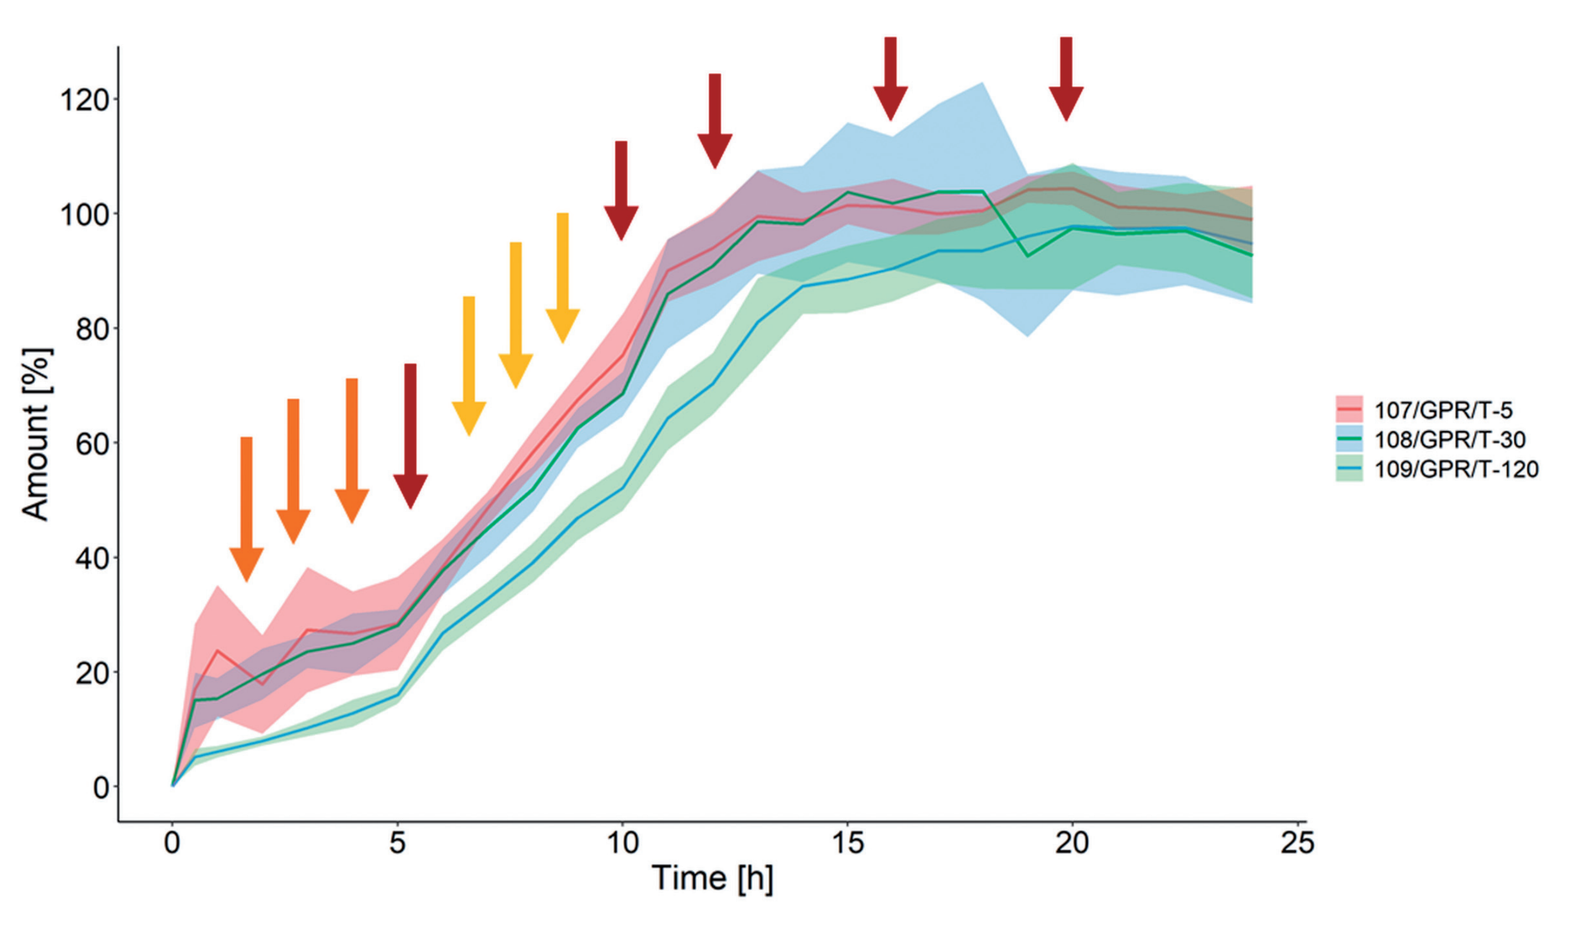
Amount [%]
Time [h]
107/GPR/T-5
108/GPR/T-30
109/GPR/T-120

Fig. 6: Dissolution profiles of three tablet strengths analyzed in stress test device under simulated fed conditions (means of n=6±SD).

Materials and Methods

CPL207-280-51 – innovative BCS class 1 API was formulated as SR matrix tablets using common excipients of pharmaceutical grade. The labelled dose amounted to 5, 30 and 120 *mg*, respectively.

Manufacturing: the tablets of three tablet strengths (5, 30, and 120 mg) were prepared on single-punch tablet press from Korsch XP1 at two different compression forces of 17 and 22 *kN*.

Standard dissolution: tests were performed using USP II paddle apparatus in 900 *mL* of phosphate buffer *pH* = 6.8 at 75 *rpm* and 37°C.

Biopredictive dissolution test: estimation of the drug release under simulated fasted and fed conditions was performed using the bio-relevant dissolution stress test device developed by Physiolution GmbH (Fig. 1). The device mimics the mechanical and physicochemical conditions of the GI passage in form of bio-predictive test algorithms designed for simulation of fasted and fed intake conditions of clinical trials.

Analytics: samples were analyzed by HPLC with fluorescence and UV detection.

Conclusion

During the development of SR tablet containing new GPR40 agonist for FIH clinical trial a composition of determined properties was evolved. Based on stress test device results, weak points of the tested formulations were identified. The ones of undesired properties were rejected. The final formulation ensured drug release in desired time, in accordance to clinical requirements. The formulation was proved to be resistant to GI tract conditions (e.g. pH changes, mechanical stress), which makes it relevant to use regardless dosing conditions designed in clinical study protocol. All planned tablet strengths were expected to release a studied API with similar dissolution rate. It should result in proper exposition of the drug to patients in the clinical trial as well as reduction of the potential risk related to FIH administration.

Acknowledgments

The financial supports of Polish National Centre for Research and Development (POIR.01.01.01-00-0334/17-00, POIR.01.02.00-00-0011/17-00) are kindly acknowledged. D. Danielak acknowledges the support of the European Union's Horizon 2020 research and innovation program under the Marie Skłodowska-Curie grant agreement No 778051, for travel and accommodation.

Contact

Ewelina Juszczyk, e-mail: ewelina.juszczyk@celonpharma.com
website: https://celonpharma.com

Referemces

1. Garbacz et al. (2010). Expert Opin. Drug Deliv. 7(11).
2. Kulinowski et al. (2016). Int J Pharm 499(1-2).